Rationale for Edasalonexent Dose Schedule in Phase 2 of the MoveDMD® Trial

H. Liu[1], R.S. Finkel[2], H.L. Sweeney[3], K. Vandenborne[3], M. Mancini[1], J.F. Reilly[1], P. Bista[1], D. Wachtel[1], S. Chandran[1], R. Dagher[1], D. Picarella[1], A. Fretzen[1], A. Nichols[1], J. Donovan[1]

[1]Catabasis Pharmaceuticals, Cambridge, MA, US; [2]Nemours Children's Hospital, Orlando, FL, US; [3]University of Florida, Gainesville, FL, US

Background

Duchenne muscular dystrophy (DMD) is a progressively debilitating and ultimately fatal inherited neuromuscular disorder caused by mutations in the gene encoding dystrophin. NF-kB is activated in DMD and drives inflammation, fibrosis, and muscle degeneration, while suppressing muscle regeneration. Edasalonexent is an oral small molecule that inhibits NF-κB. In the Phase 2 and the open-label extension (OLE) portions of the MoveDMD trial enrolling 4- to 7-year-old boys with DMD, edasalonexent given 33 mg/kg three times daily (TID) (100 mg/kg/day) demonstrated a preservation of muscle function and slowing of DMD disease progression through 60 weeks as compared to the rates of change during the control period prior to edasalonexent treatment.

The MoveDMD trial was designed to evaluate efficacy, safety/tolerability, pharmacokinetics (PK), pharmacodynamics (PD), and dose response of edasalonexent. The Phase 1 portion of the MoveDMD trial was a 1-week study to evaluate the safety, PK, and PD at 33 and 67 mg/kg/day, given in 2 divided doses, and 100 mg/kg/day given in 3 divided doses. Phase 2 was a 12-week placebo-controlled study with two cohorts given 33 mg/kg twice daily (BID) (67 mg/kg/day) and 33 mg/kg TID (100 mg/kg/day). Patients in the OLE continued with either dose regimen of edasalonexent.

The dose schedule for the Phase 2 of the MoveDMD trial was selected based on nonclinical and clinical data including (1) the exposure/efficacy relationship observed in animal models; (2) the Phase 1 safety, tolerability, and PD in pediatric DMD patients; and (3) human PK.

In the *mdx* mouse model of DMD, dose efficacy was driven by sustained systemic exposure. An oral dose of edasalonexent given TID showed greater pharmacological effects than the same dose given all at once daily despite similar AUC.

Sustained Edasalonexent Exposure in *mdx* Mice Reduces Inflammation & Fibrosis

- **Administration of edasalonexent in diet resulted in sustained plasma exposure and reduced muscle inflammation and fibrosis in *mdx* mice**
- **Short-term high exposure of edasalonexent by once-daily (QD, 450 mg/kg) oral gavage is not efficacious; combination of QD oral dosing and diet administration is no more effective than diet alone**

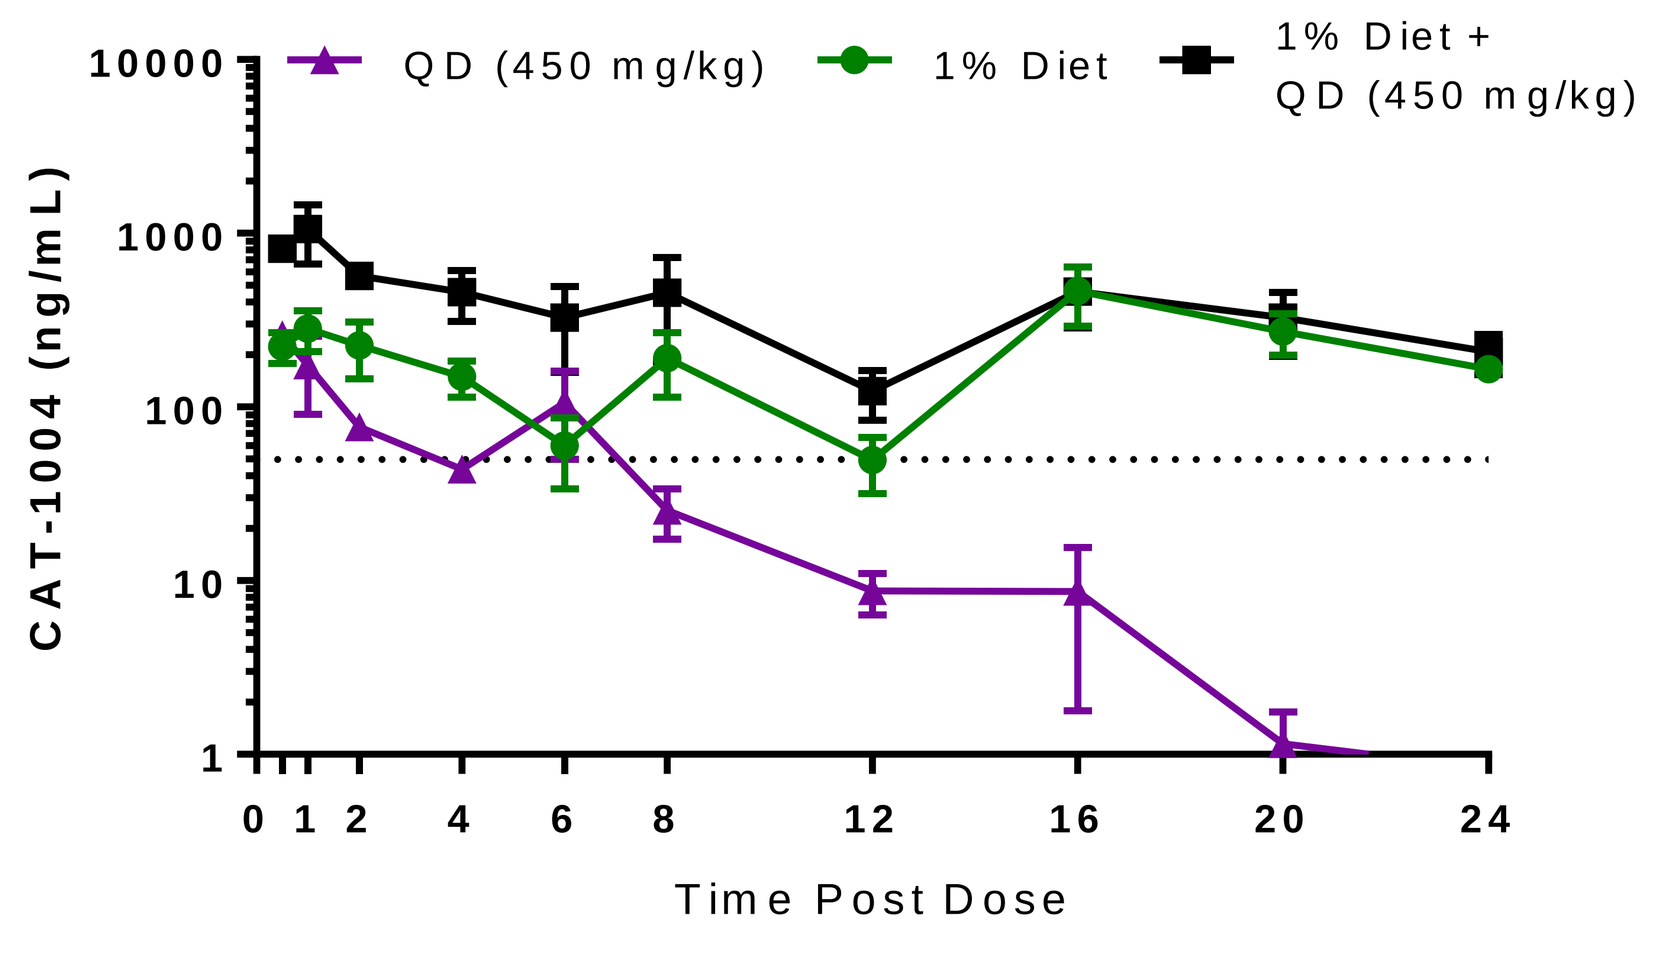
QD (450 mg/kg)
1% Diet
1% Diet + QD (450 mg/kg)
CAT-1004 (ng/mL)
Time Post Dose

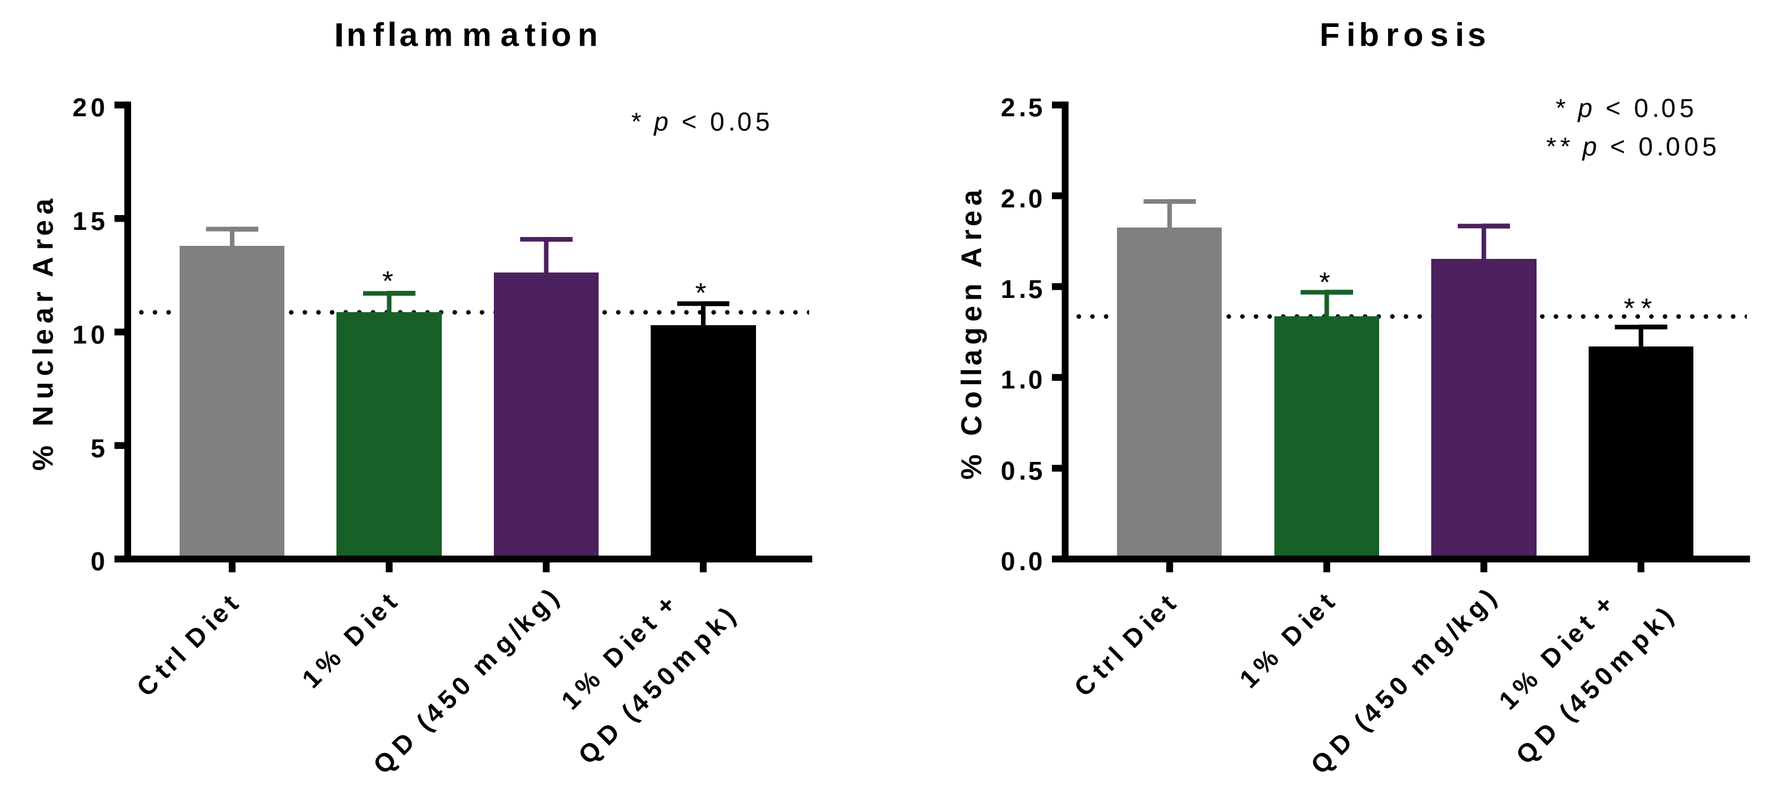
Inflammation
* p < 0.05
% Nuclear Area
Fibrosis
* p < 0.05
** p < 0.005
% Collagen Area
Ctrl Diet
1% Diet
QD (450 mg/kg)
1% Diet + QD (450mpk)

Sustained Edasa Exposure in DMD Patients Correlates with NF- κB Inhibition

- **In DMD patients, systemic exposures achieved levels at which NF-κB inhibition was observed in adults**
 - In a previous Phase 1 study in adults, changes in expression of NF-κB driven genes were observed in whole blood at a dose of approximately 100 mg/kg/day
 - In the MoveDMD study, when doses of 33 mg/kg were given BID or TID (total daily doses of 67 or 100 mg/kg), systemic exposures reached levels at which NF-κB inhibition was observed in adults
 - Exposures consistent with efficient release of DHA observed in preclinical studies

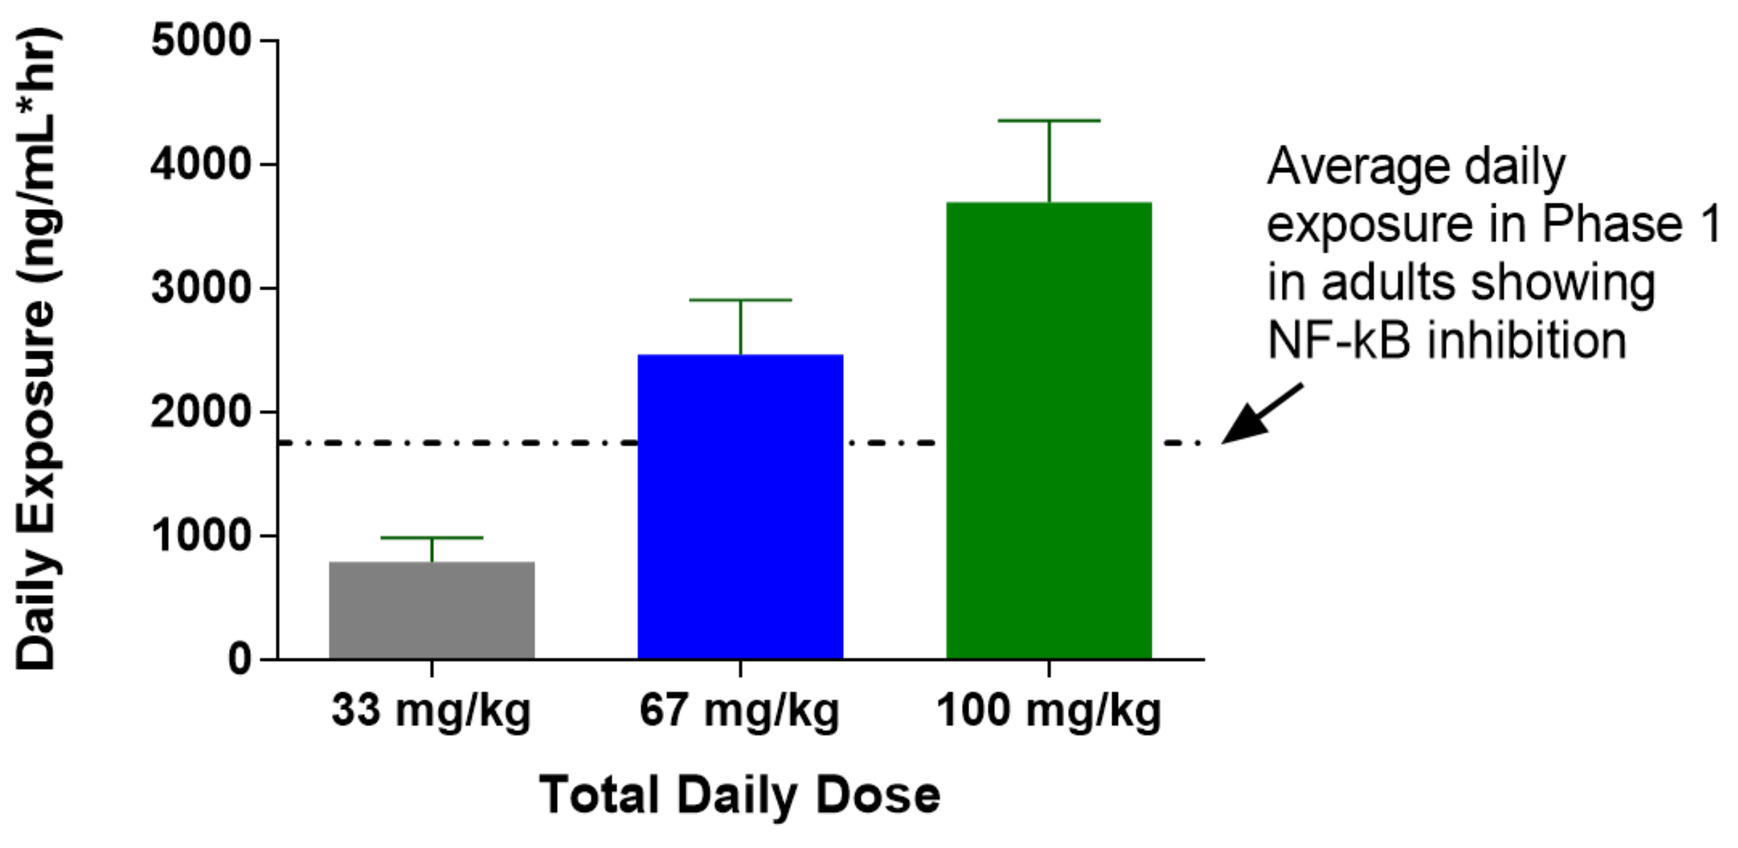
Daily Exposure (ng/mL*hr)
Average daily exposure in Phase 1 in adults showing NF-kB inhibition
33 mg/kg
67 mg/kg
100 mg/kg
Total Daily Dose

- **TID dosing substantially increases time over threshold compared to BID dosing**

Plasma concentration after 1st 33 mg/kg oral dose (Phase 2 and Open Label Extension)

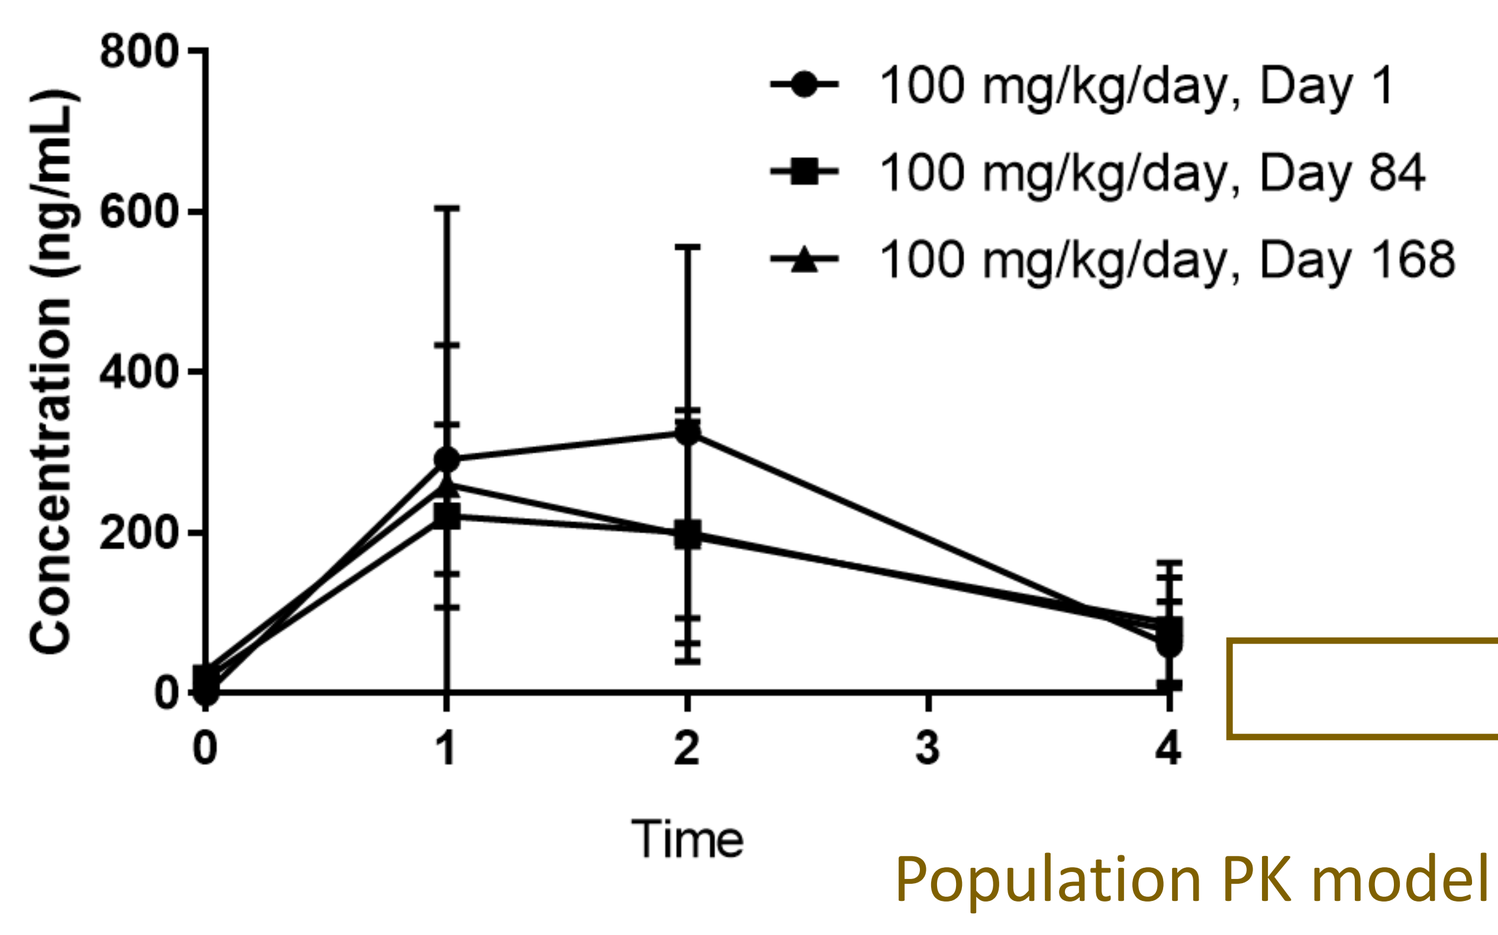
100 mg/kg/day, Day 1
100 mg/kg/day, Day 84
100 mg/kg/day, Day 168
Concentration (ng/mL)
Time

Population PK model developed using PK data from MoveDMD trial and Phase 1 PK data in adults

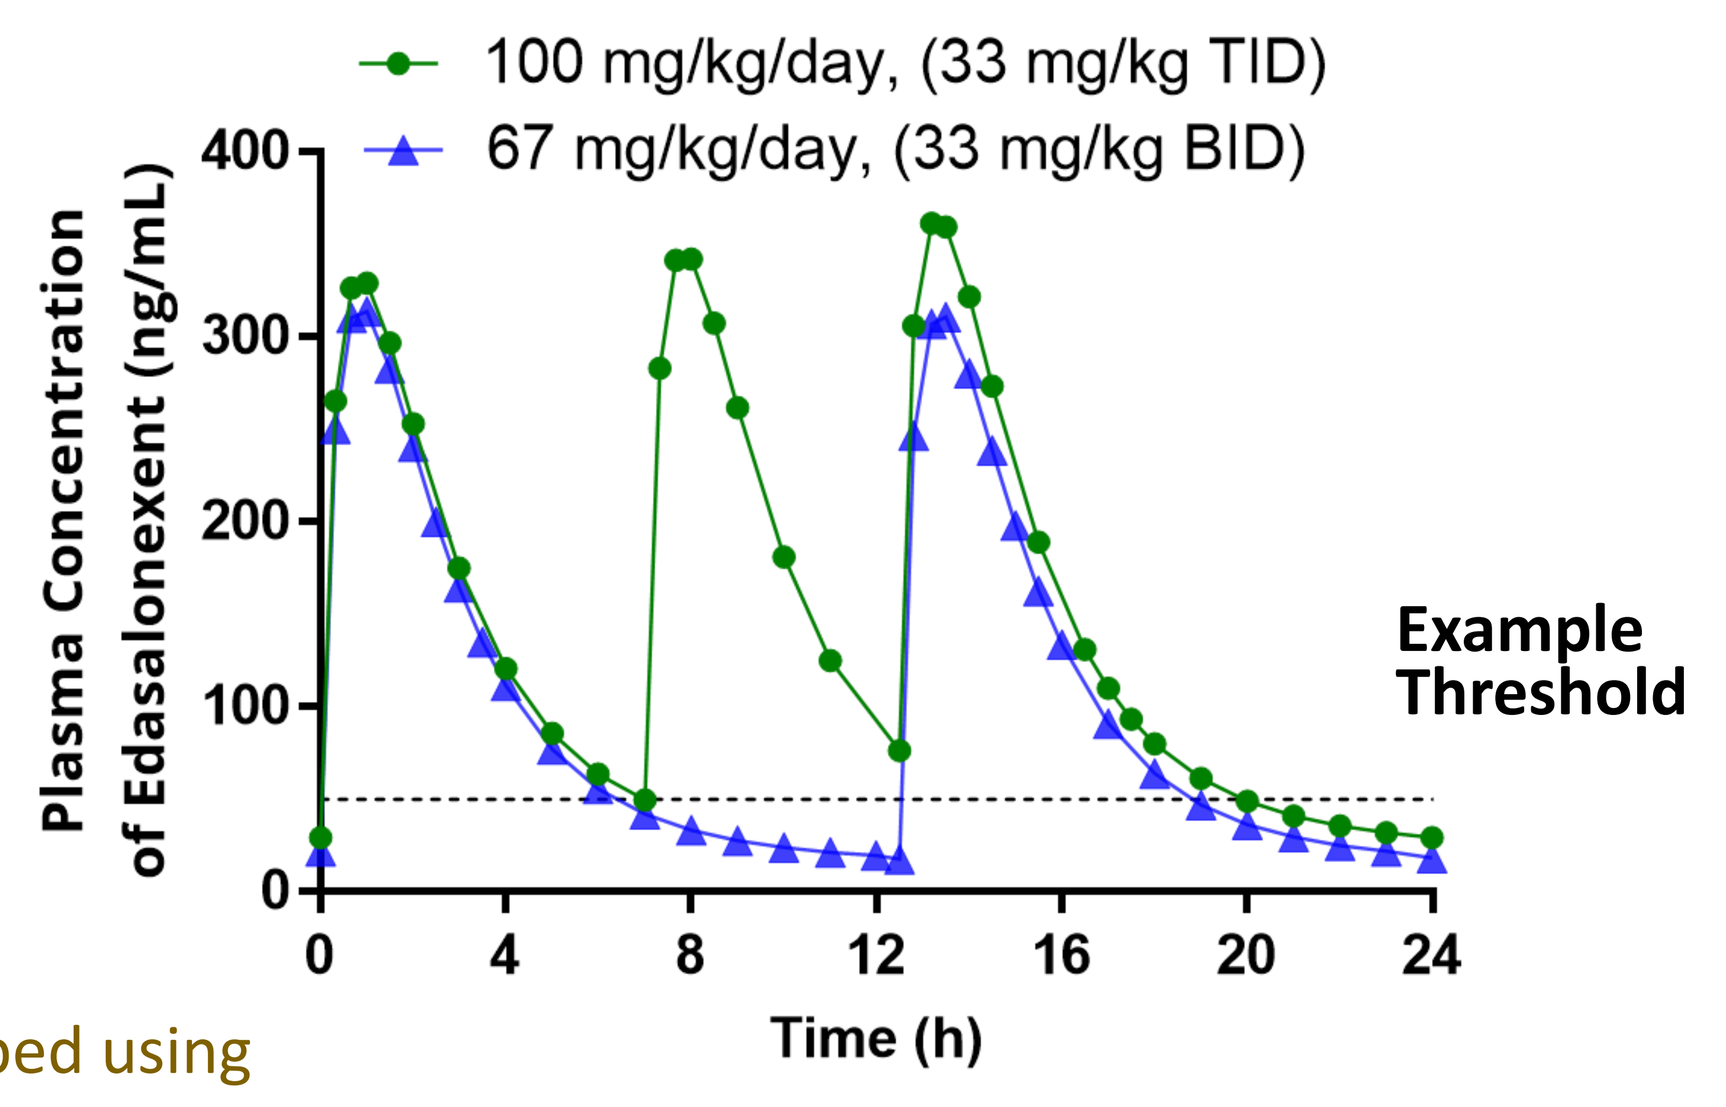
100 mg/kg/day, (33 mg/kg TID)
67 mg/kg/day, (33 mg/kg BID)
Plasma Concentration of Edasalonexent (ng/mL)
Example Threshold
Time (h)

- **Edasalonexent produces dose-related reductions in NF-κB regulated and inflammation-related gene transcripts in whole blood of DMD patients**

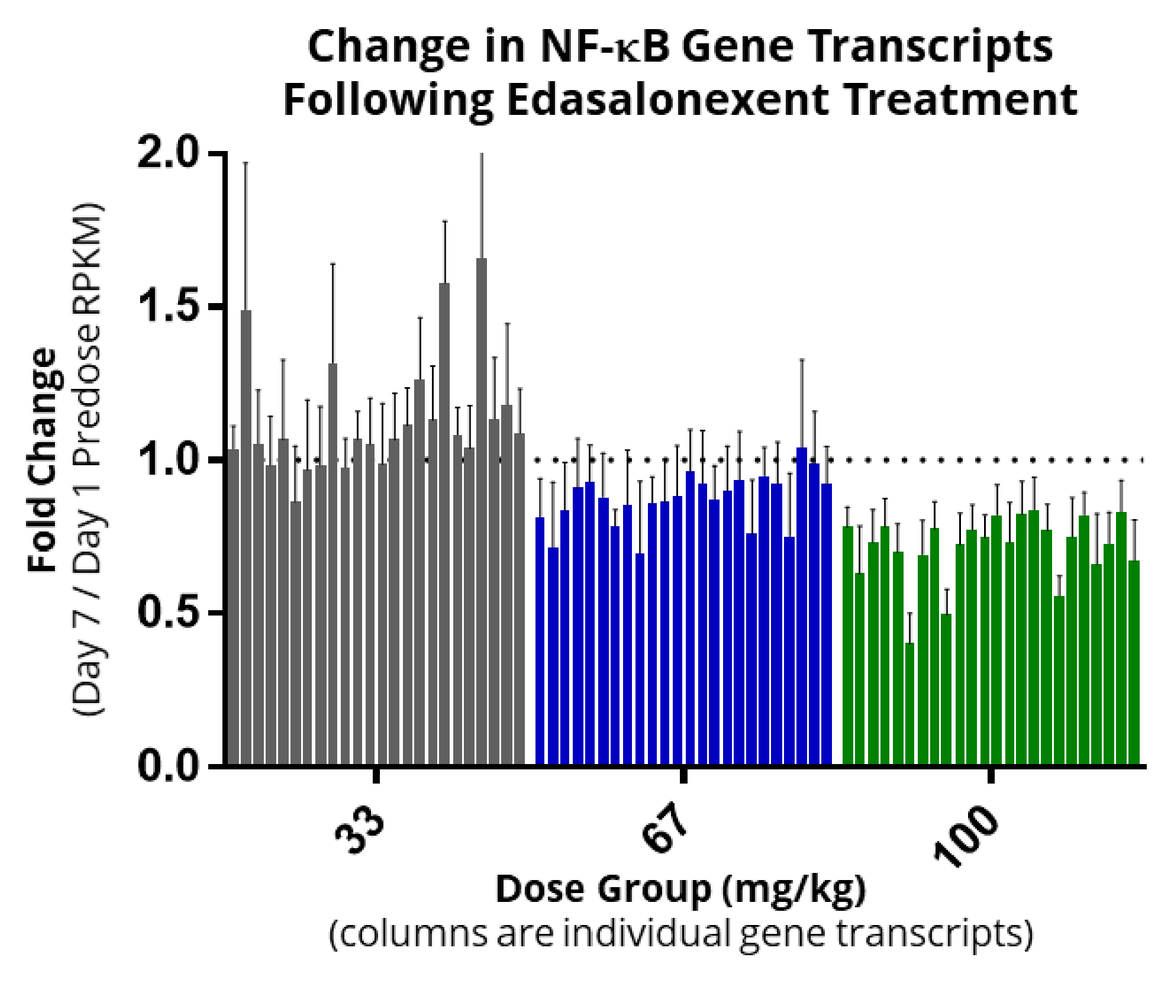
Change in NF-κB Gene Transcripts
Following Edasalonexent Treatment
Fold Change
(Day 7 / Day 1 Predose RPKM)
Dose Group (mg/kg)
(columns are individual gene transcripts)

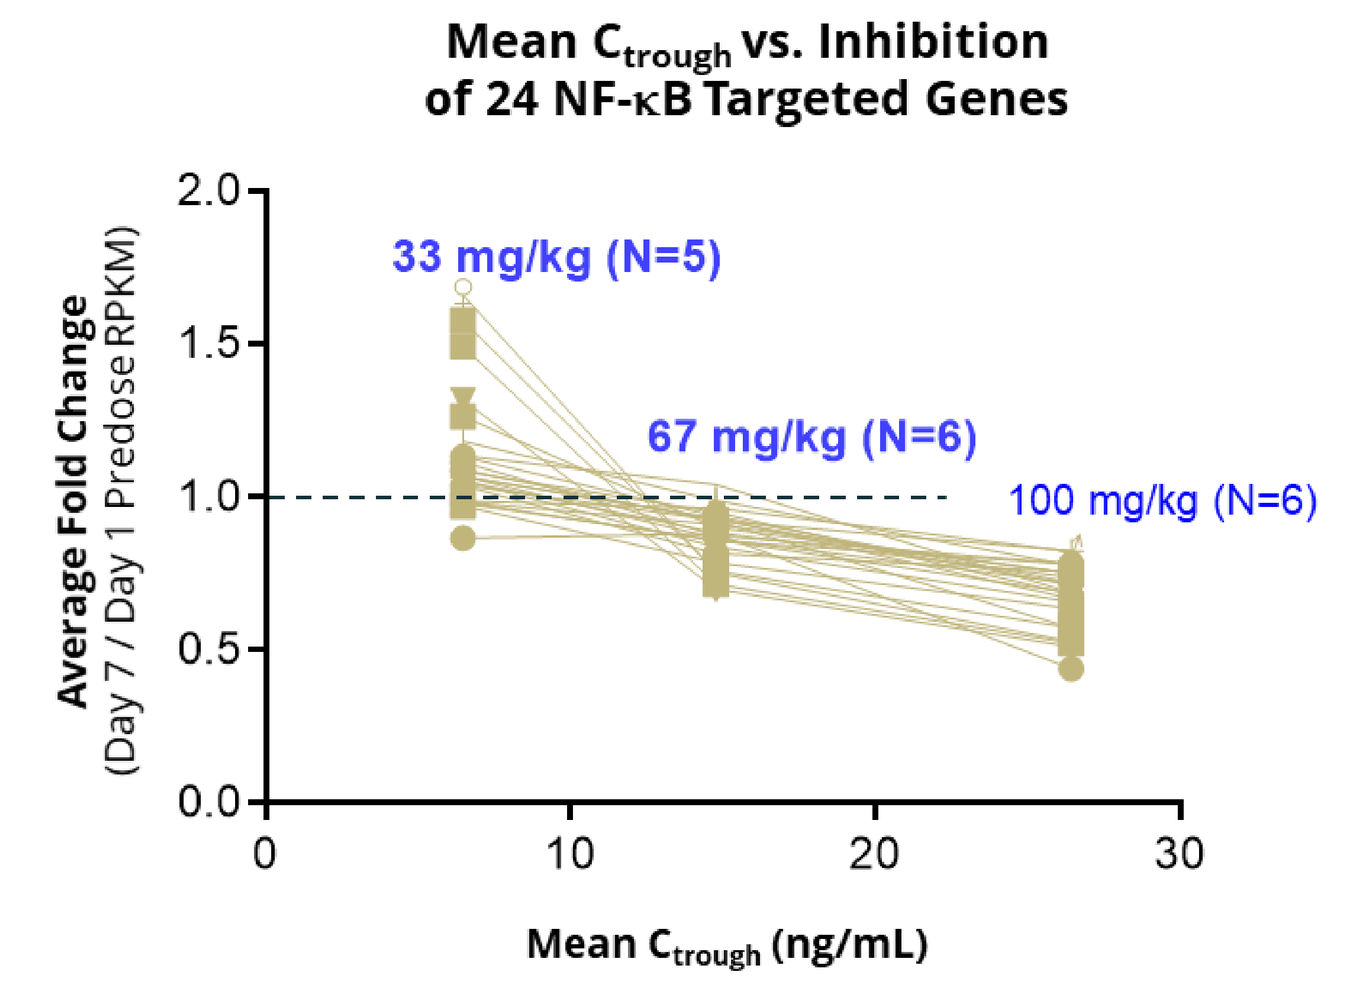
Mean Ctrough vs. Inhibition
of 24 NF-κB Targeted Genes
33 mg/kg (N=5)
67 mg/kg (N=6)
100 mg/kg (N=6)
Average Fold Change
(Day 7 / Day 1 Predose RPKM)
Mean Ctrough (ng/mL)

- C_{trough} is a driver of efficacy in preclinical models and in the clinic
- Increasing time over threshold with TID dosing produces pharmacodynamic effects

Background: MoveDMD Trial Design

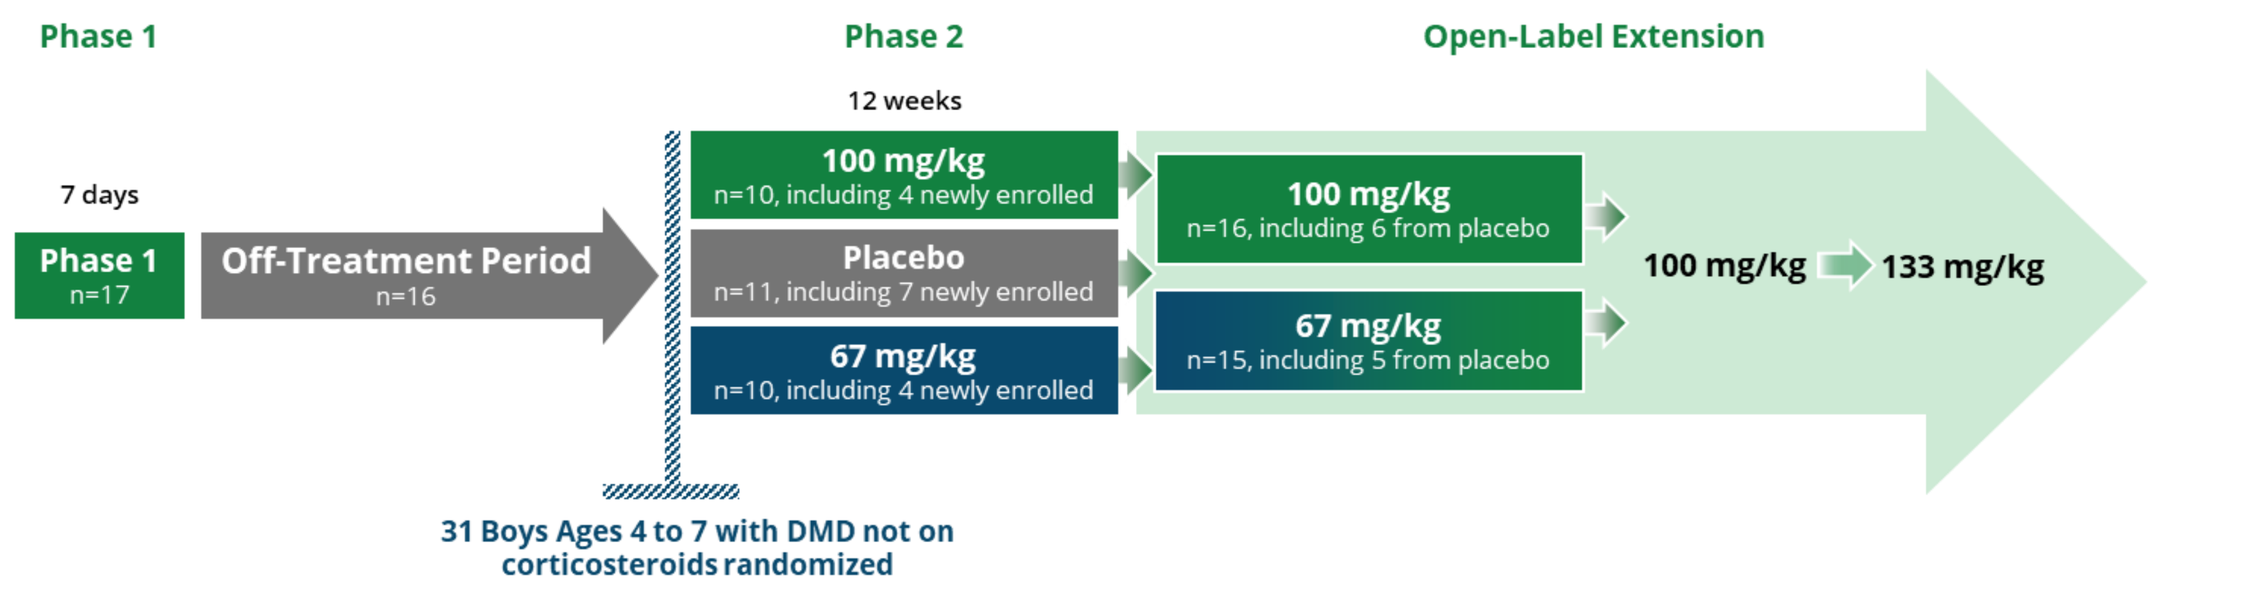
Phase 1
Phase 2
Open-Label Extension
12 weeks
7 days
Phase 1
n=17
Off-Treatment Period
n=16
100 mg/kg
n=10, including 4 newly enrolled
Placebo
n=11, including 7 newly enrolled
67 mg/kg
n=10, including 4 newly enrolled
100 mg/kg
n=16, including 6 from placebo
67 mg/kg
n=15, including 5 from placebo
100 mg/kg
133 mg/kg
31 Boys Ages 4 to 7 with DMD not on corticosteroids randomized

- **Integrated multi-part trial design**
- **Off-treatment control period measurements between Phase 1 and commencement of dosing in Phase 2 and open-label extension**
 - Provides internal control for pre-specified MoveDMD analyses
- **Open-label extension**

Edasalonexent Treatment Stabilizes NSAA and Other Functional Measures

- **North Star Ambulatory Assessment Score (NSAA) is a measure of overall function in young boys**
- **Disease Progression (as measured by NSAA) on edasalonexent improved compared with rate of change during off-treatment control period**
- **All Timed Function Tests stabilized with edasalonexent treatment compared with rate of change during off-treatment control period, consistent with effect on NSAA**

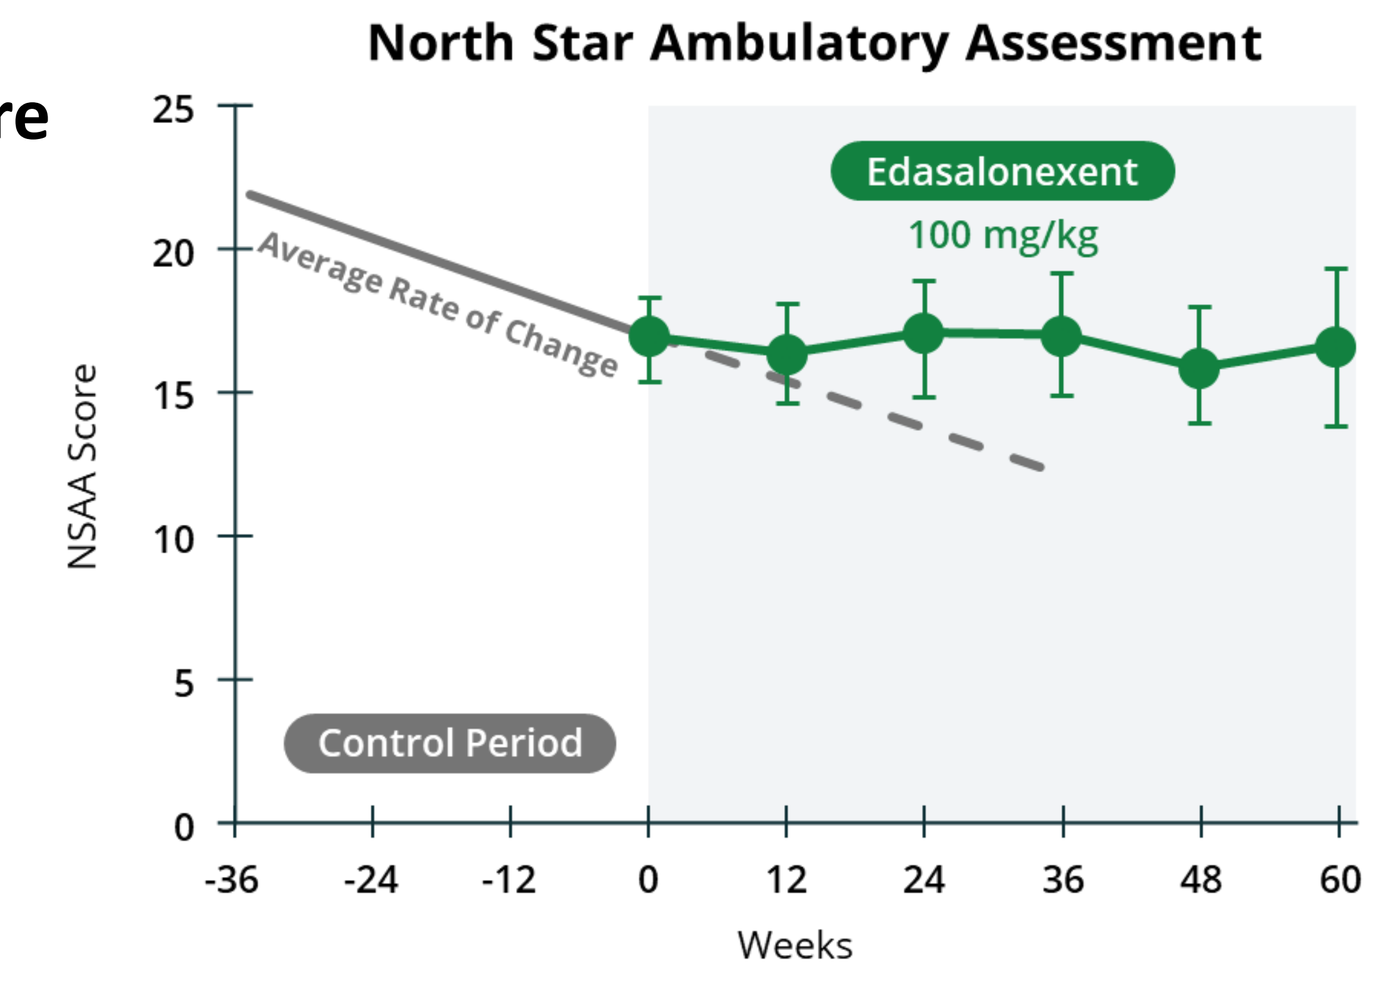
North Star Ambulatory Assessment
Edasalonexent
100 mg/kg
Average Rate of Change
NSAA Score
Control Period
Weeks

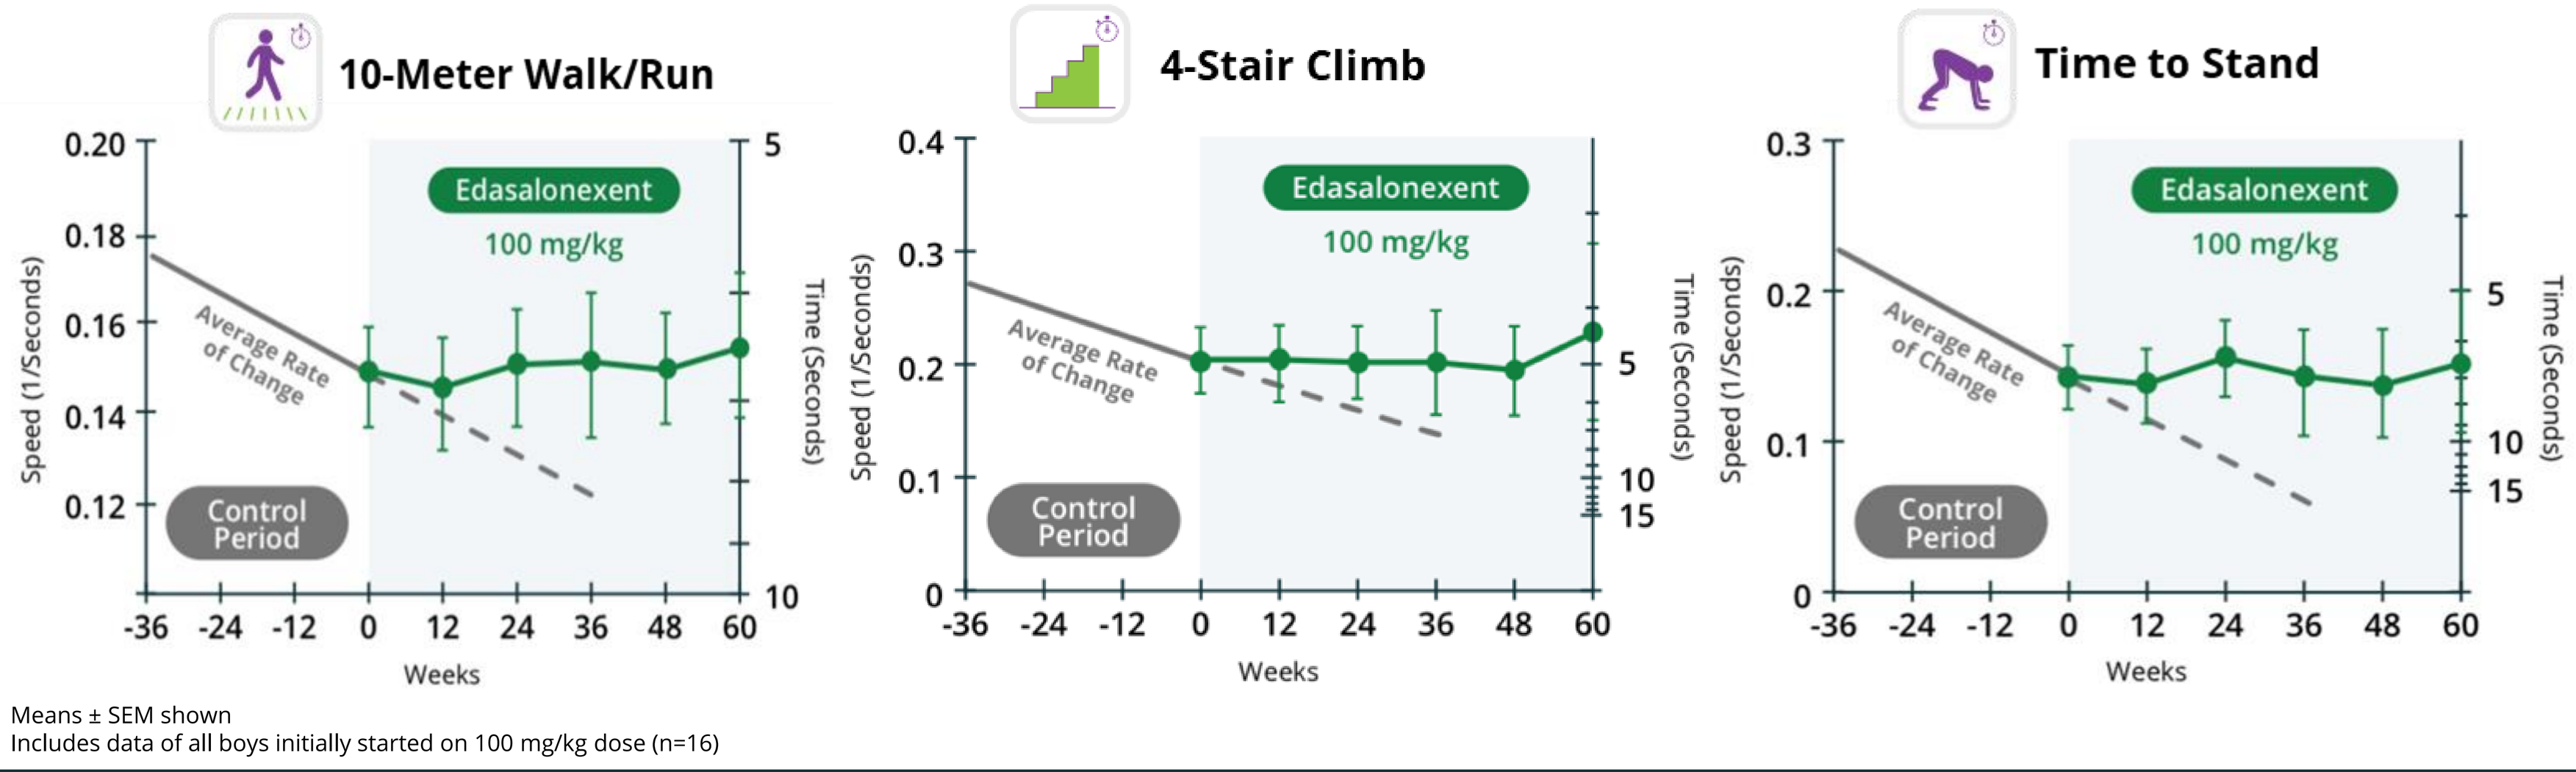
10-Meter Walk/Run
4-Stair Climb
Time to Stand
Edasalonexent
100 mg/kg
Average Rate of Change
Control Period
Speed (1/Seconds)
Time (Seconds)
Weeks

Means ± SEM shown
Includes data of all boys initially started on 100 mg/kg dose (n=16)

Edasalonexent Significantly Improved Rate of Change of MRI T2

- **MRI T2 and MRS Fat Fraction are non-invasive measures of disease progression in DMD that are elevated and increase with age in DMD**
- **MoveDMD incorporated both MRI and MRS**
- **Changes in MRI T2 and Fat Fraction strongly correlate with changes in function and predict future loss of functional milestones** *(Willcocks et al, 2016, Ann. Neurol., Willcocks et al, 2014, Ann. Neuro; Barnard et al. 2018 PLoS)*

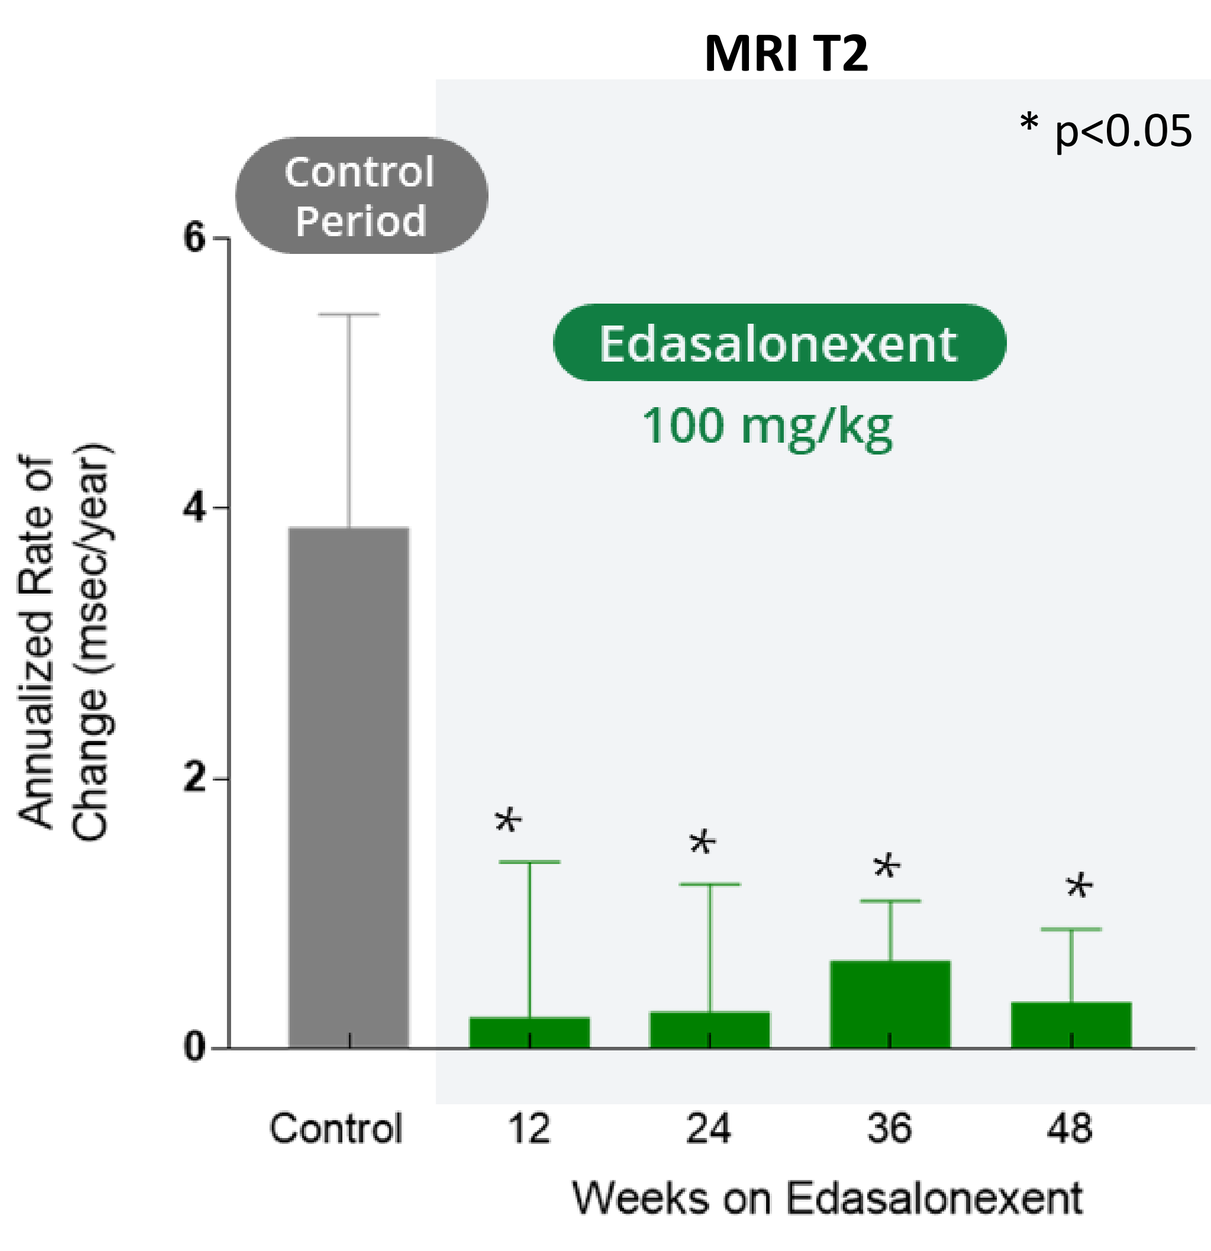
MRI T2
* p<0.05
Control Period
Edasalonexent
100 mg/kg
Annualized Rate of Change (msec/year)
Control
Weeks on Edasalonexent

MR Spectroscopy
Change in Fat Fraction from Baseline

Muscle	MoveDMD Off-Treatment Control Period Annualized Rate	MoveDMD 48 weeks on Edasalonexent	ImagingDMD Natural History Study* 1 Year Change
Soleus	2.6%	0.85%	3%
Vastus lateralis	10.4%	5.9%	7%

-In ImagingDMD study, boys were largely on chronic steroids (>75%)

- **Edasalonexent significantly improved rate of change of MRI T2 (composite of 5 lower leg muscles)** compared to the off-treatment period
 - Stabilization of MRI T2 is consistent with slowing of disease progression also observed in functional assessments
- **Edasalonexent decreased the rate of increase in fat fraction of the soleus (calf) and vastus lateralis (a quadriceps) compared to the off-treatment period**

Edasalonexent was Well Tolerated with No Safety Signals

- **No safety signals in MoveDMD trial to date**
- **Well tolerated, with majority of adverse events being mild in nature, mostly gastrointestinal**
- **No adverse trends in hematology, chemistry, renal or adrenal function, calcium and phosphate**
- **Growth: age appropriate increases in weight and height**
- **ECG heart rate decreased toward age-normative values**

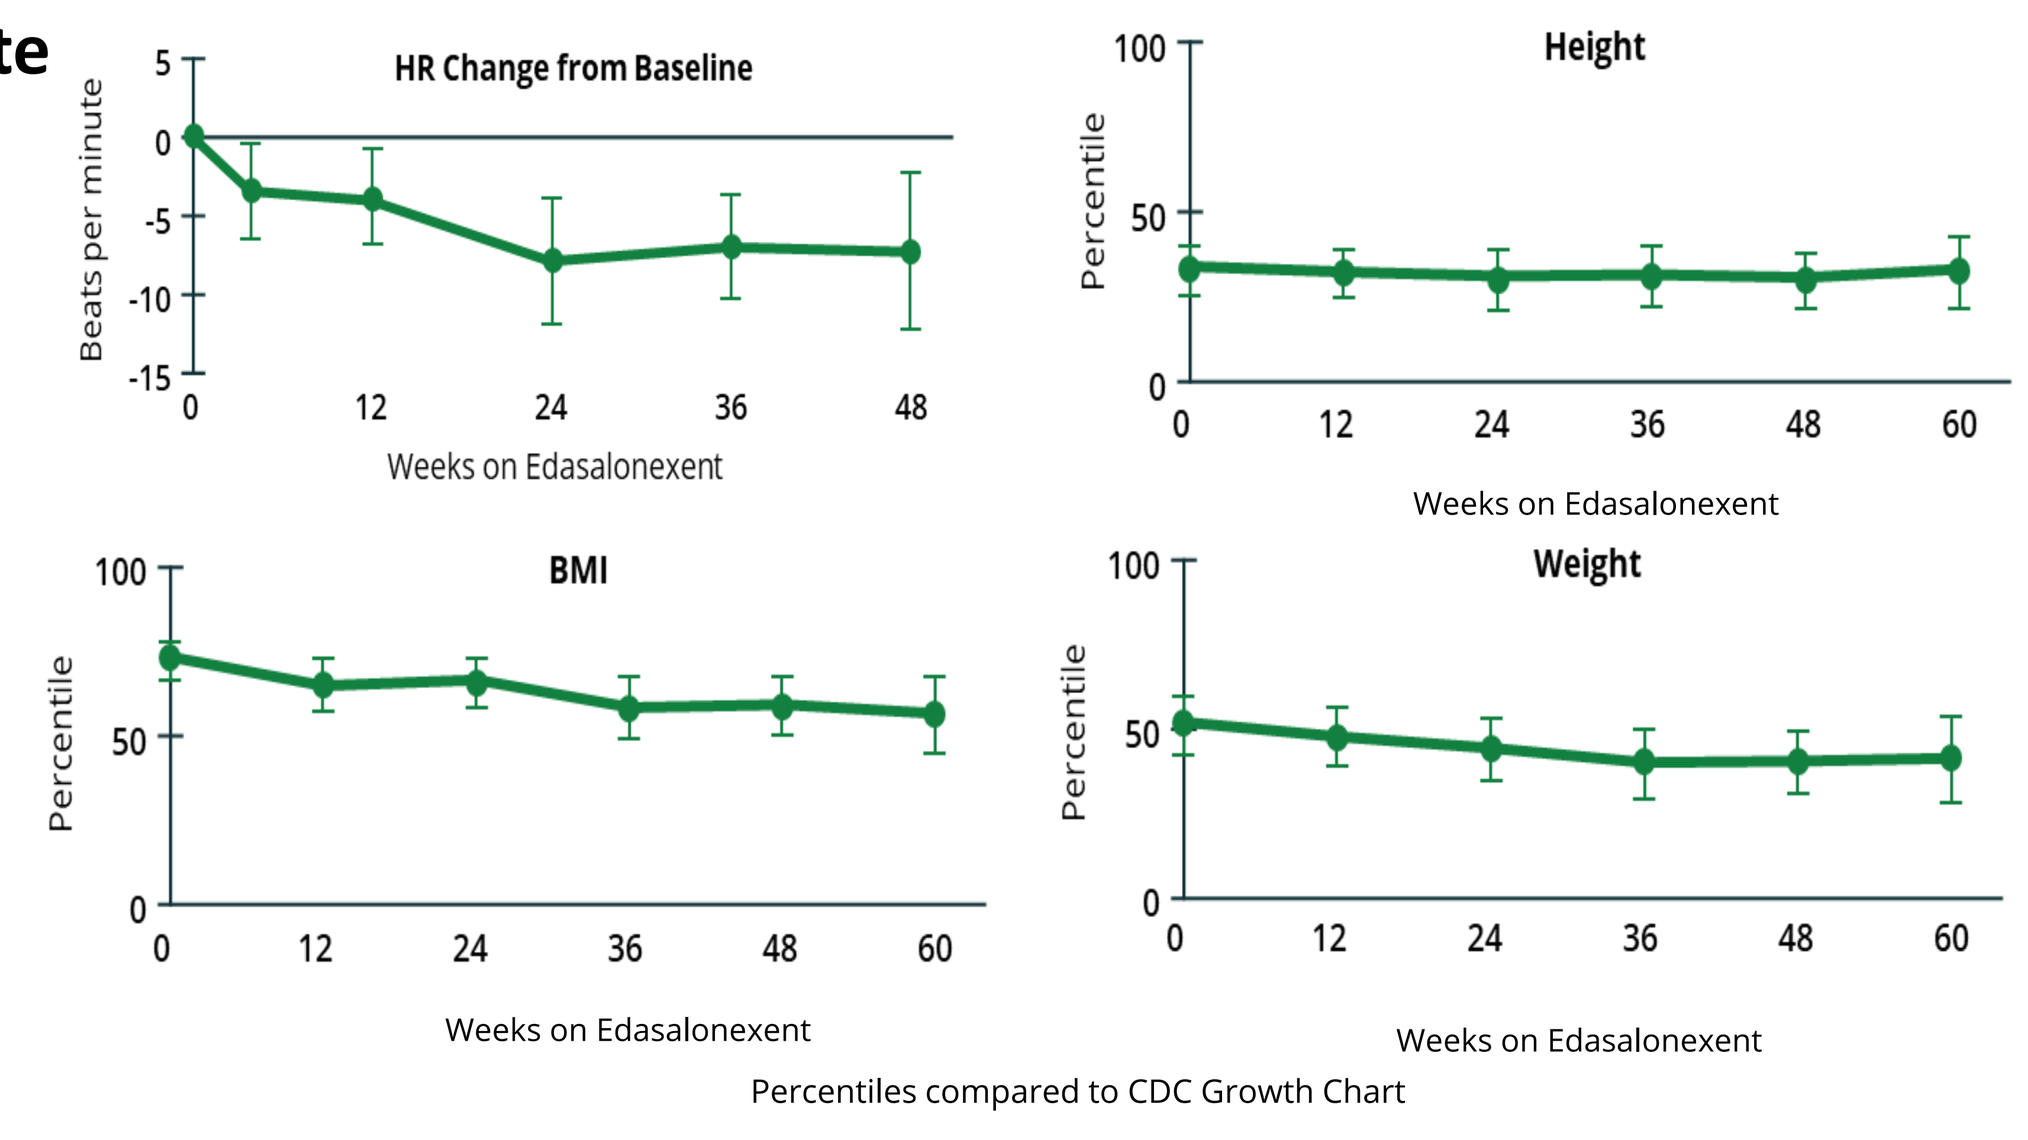
HR Change from Baseline
Beats per minute
Height
BMI
Weight
Percentile
Weeks on Edasalonexent
Percentiles compared to CDC Growth Chart

Summary and Conclusions

- **Results from preclinical studies and systemic exposures in DMD patients suggest sustained exposure and time over threshold are drivers for pharmacodynamic signal and efficacy**
- **Population PK model from MoveDMD trial suggests sustained exposure above a threshold level can be achieved with TID dose schedule of edasalonexent**
- **TID dosing (100 mg/kg/day) in MoveDMD trial and open-label extension show clinically meaningful slowing of disease progression over >1 year compared to off-treatment control period**
- **MRI measures support positive edasalonexent treatment effects over 48 weeks**
- **There were no safety signals with edasalonexent treatment over >1 year**
- **These results support upcoming Phase 3 clinical trial in DMD**

Abstract # 753

Presented at the 15th International Congress on Neuromuscular Diseases (ICNMD) | July 6-10, 2018| Vienna, Austria