Edasalonexent, a Novel NF-kB Inhibitor, Slows Disease Progression in Young Boys with Duchenne in the Phase 2 and Open-Label Extension of MoveDMD Trial

Richard Finkel, MD[1], Krista Vandenborne, PT PhD[2], H. Lee Sweeney, PhD[2], Erika L. Finanger, MD[3], Gihan I. Tennekoon, MBBS MRCS LCRP[4], Perry Shieh, MD PhD[5], Rebecca J. Willcocks, PhD[2], Glenn Walter, PhD[2], William Rooney, PhD[3], Sean C. Forbes, PhD[2], William T. Triplett, BSc[2], Sabrina W. Yum, MD[4], Maria Mancini, MHP[6], James MacDougall, PhD[6],, Joanne M. Donovan, MD PhD[6]

[1] Nemours Children's Health System, Orlando, FL; [2] University of Florida Health, Gainesville, FL; [3] Oregon Health Sciences University, Portland, OR; [4] The Children's Hospital of Philadelphia, Philadelphia, PA; [5] University of California, Los Angeles, Los Angeles, CA; [6] Catabasis Pharmaceuticals, Cambridge, MA

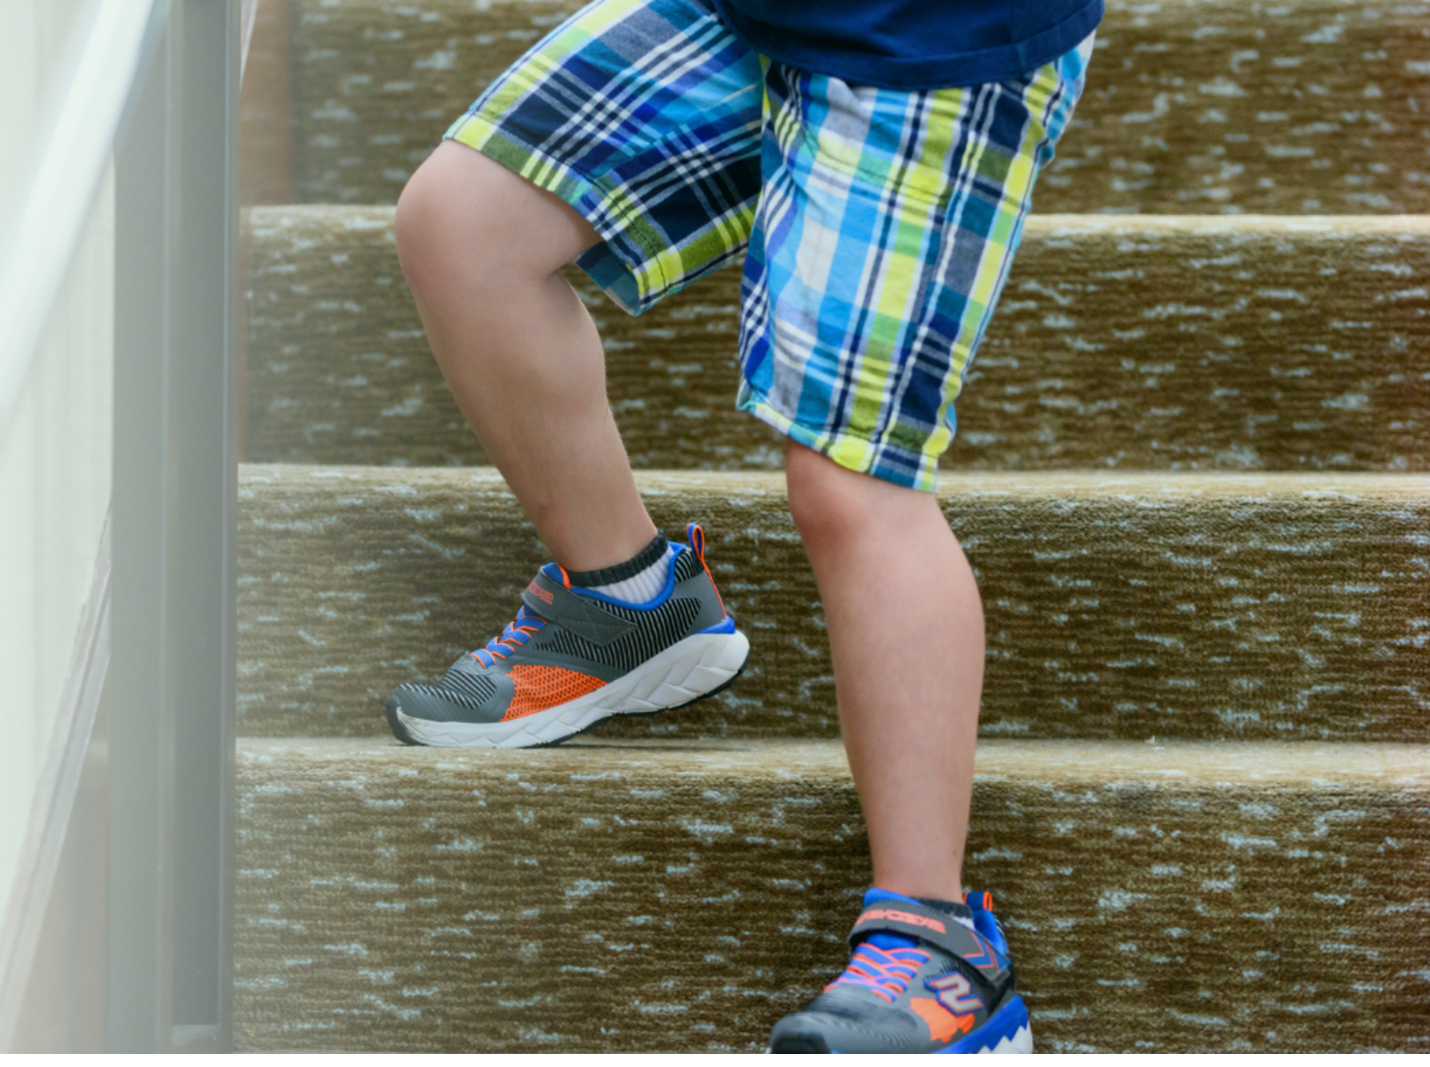

Background & Study Design

Edasalonexent: Potential for Broad Therapeutic Benefit

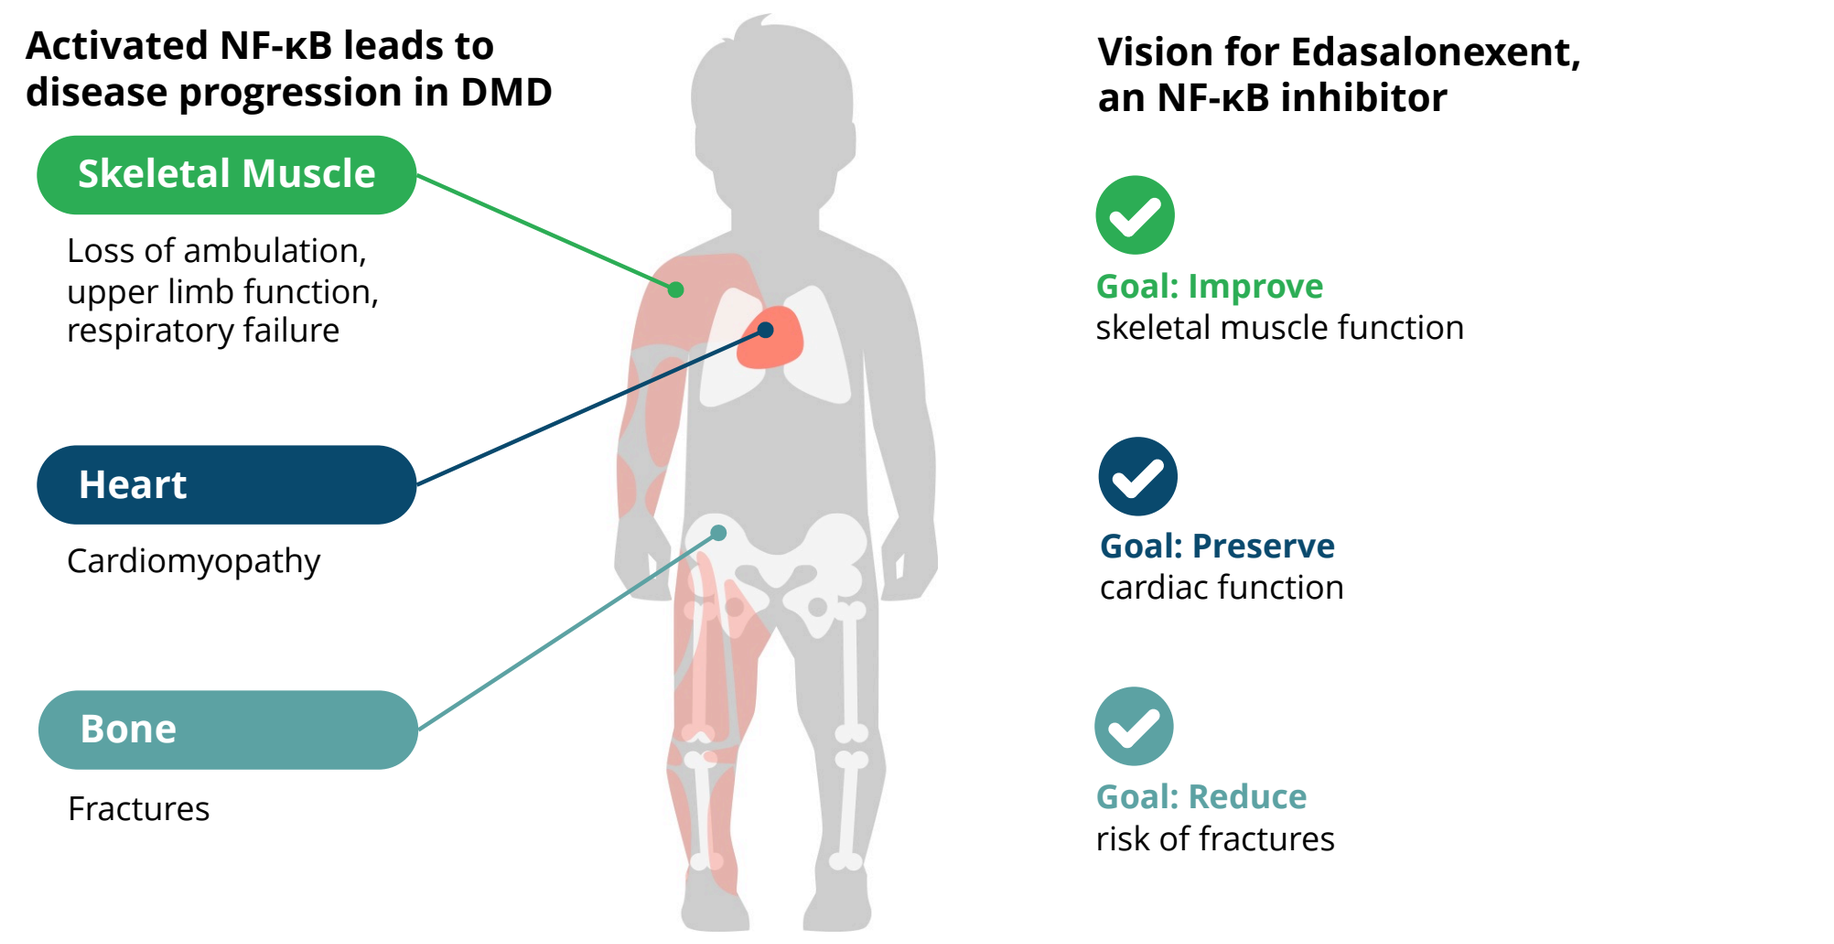

Activated NF-κB leads to disease progression in DMD
Skeletal Muscle
Loss of ambulation, upper limb function, respiratory failure
Heart
Cardiomyopathy
Bone
Fractures
Vision for Edasalonexent, an NF-κB inhibitor
Goal: Improve skeletal muscle function
Goal: Preserve cardiac function
Goal: Reduce risk of fractures

NF-κB is a key link between loss of dystrophin and disease pathology; it plays a fundamental role in the initiation and progression of skeletal muscle, respiratory and cardiac disease in DMD

Design of MoveDMD, a Phase 2 Trial with Open-Label Extension

- **Study Objectives**
 - Proof of concept using MRI to assess changes in muscle health
 - Long-term study to enable Phase 3
- **Key Inclusion / Exclusion criteria**
 - Age 4 to 7 (up to 8th birthday) not on corticosteroids for at least 24 weeks

Phase 1 n=17	Off-Treatment Period n=16	Phase 2 n=31	Open-Label Extension n=31
1 week	**~6 months** prior to Phase 2	**12-week** placebo-controlled period	**>72** weeks

- **Analysis Plan**
 - 12-week placebo control period
 - **Compare changes during off-treatment control period with changes after initiation of edasalonexent**

Key Functional Assessments Performed During Clinic Visits

North Star Ambulatory Assessment

Assessment measures — from most to least difficult

Hop right leg	Climb box step right
Hop left leg	Climb box step left
Stand on heels	Stand on one leg right
Rise from floor	Stand on one leg left
Run	Get to sitting
Jump	Rise from chair
Lift head	Walk
Descend box step right	Stand
Descend box step left	

How measures are scored: 2 Can perform; 1 Can perform with difficulty; 0 Unable to perform

Timed Function Tests

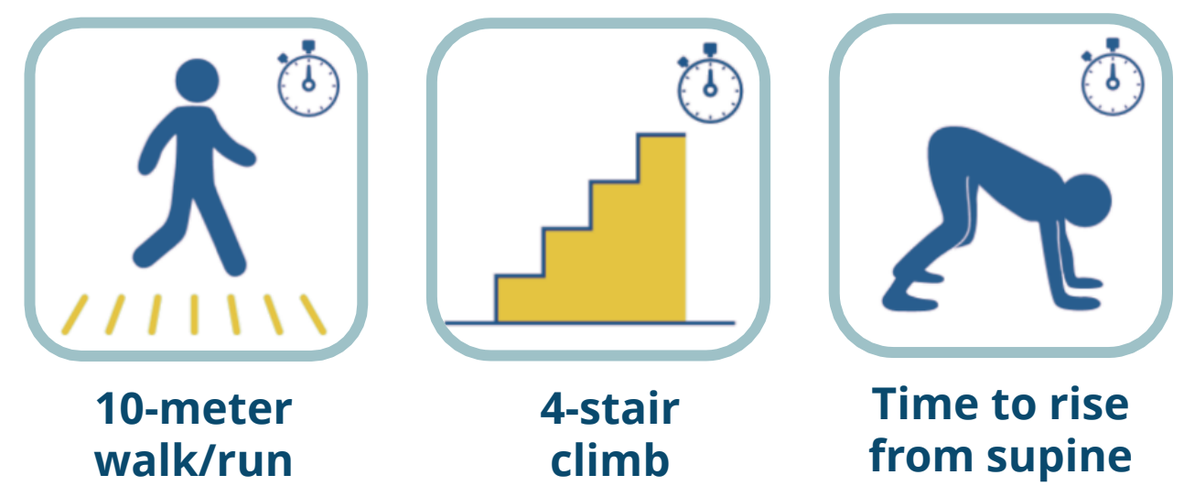

10-meter walk/run
4-stair climb
Time to rise from supine

Range of Endpoints to Demonstrate Proof of Concept and Support Design of Phase 3

NF-κB Target Engagement	Biomarkers	Muscle MRI	Functional
Inhibition of NF-κB targeted gene set in peripheral blood	CRP, biomarker of inflammation; Muscle enzymes	MRI T2 of upper and lower leg; MRS muscle fat	North Star Ambulatory Assessment and Timed Function Tests

Results

In Phase 2 MoveDMD Trial and Open-Label Extension: All Assessments of Function Stabilized on Edasalonexent Compared to Off-Treatment Control

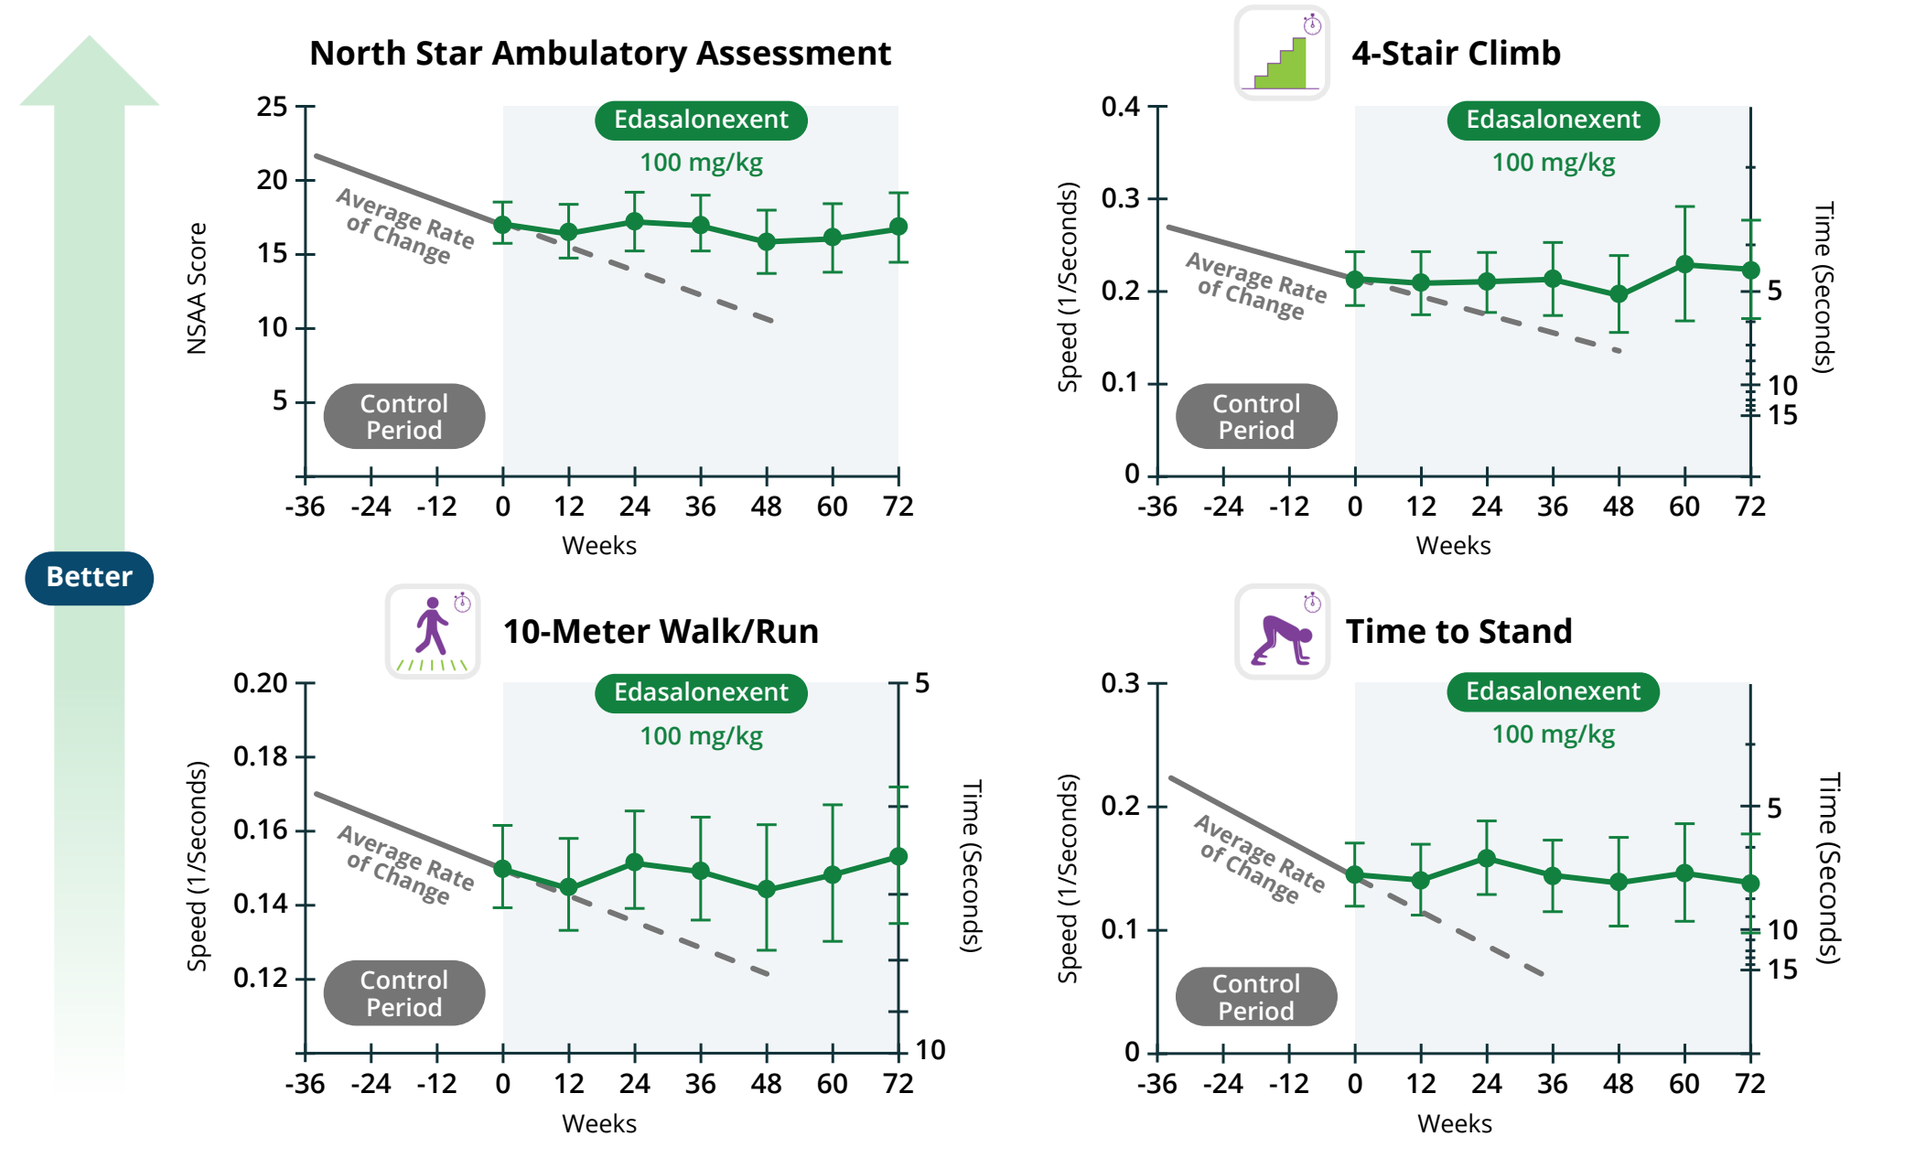

Better
North Star Ambulatory Assessment
NSAA Score
Edasalonexent 100 mg/kg
Average Rate of Change
Control Period
Weeks
4-Stair Climb
Speed (1/Seconds)
Time (Seconds)
10-Meter Walk/Run
Time to Stand

Means ± SEM shown. Includes data of all boys initially started on 100 mg/kg dose (n=16) with 11 boys participating through 72 weeks.

In Phase 2 MoveDMD Trial and Open-Label Extension: Edasalonexent Improved Rate of Change of MRI T2 Compared to Off-Treatment Control Period

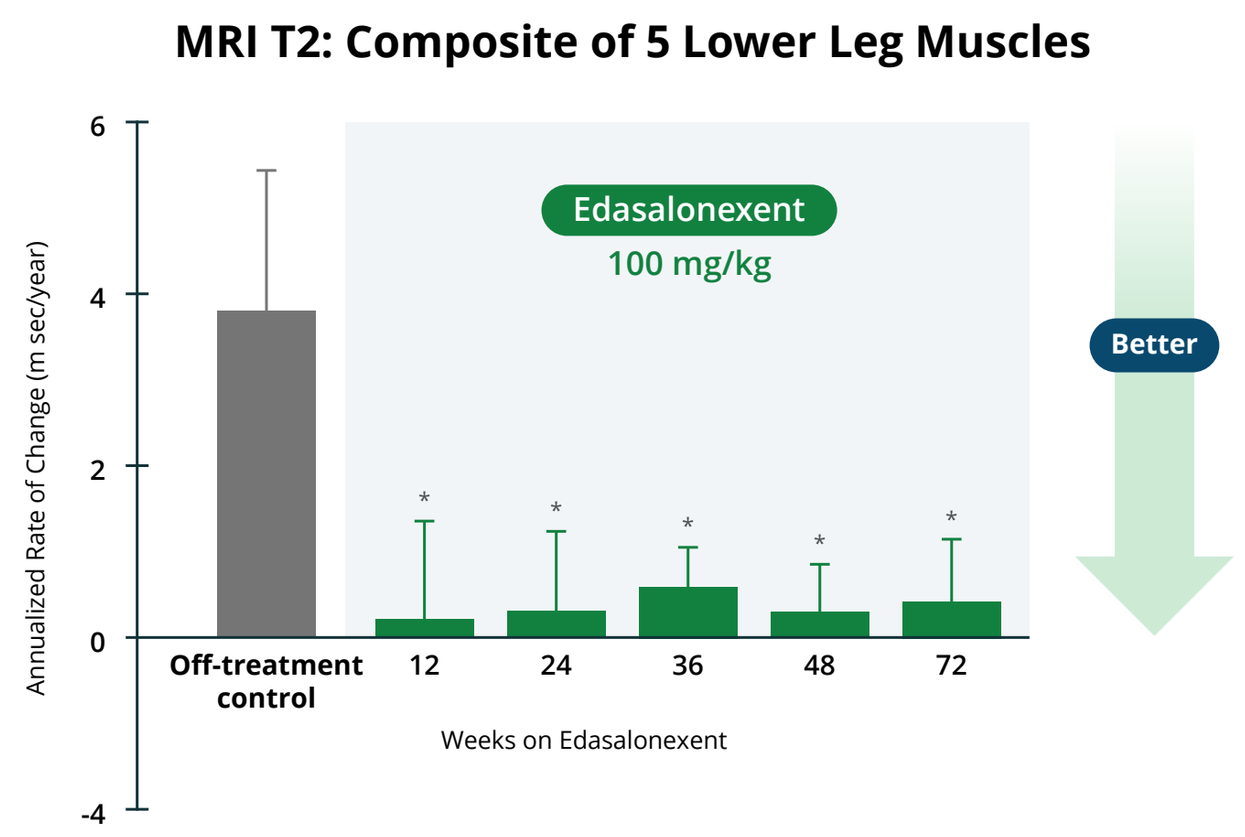

MRI T2: Composite of 5 Lower Leg Muscles
Annualized Rate of Change (m sec/year)
Edasalonexent 100 mg/kg
Better
Off-treatment control
12
24
36
48
72
Weeks on Edasalonexent

- Composite of 5 lower leg muscles MRI T2 used to encompass muscles at various stages of disease progression and minimize variability
- Following 72 weeks of edasalonexent, the rate of increase in the composite MRI T2 in the five lower leg muscles decreased as compared to the rate of increase during the off-treatment control period

Means + SEM; mixed model comparison with off-treatment period
* Week 12: p=0.002, n=16; Week 24: p=0.004, n=14; Week 36: p=0.032, n=13; Week 48: p=0.018, n=12; Week 72: p=0.052, n=9

Safety: Edasalonexent Has Been Well Tolerated To Date

- **50+ years of patient exposure**
 - Majority of adverse events observed were mild in nature
- **Boys on edasalonexent in our Phase 2 clinical trial and open-label extension grew similarly to boys not affected by Duchenne**
 - Height increased by an average of 2.1 inches/year
 - Weight increased by an average of 2.9 pounds/year
 - Both increases in line with typical height and weight increases of boys not affected by Duchenne

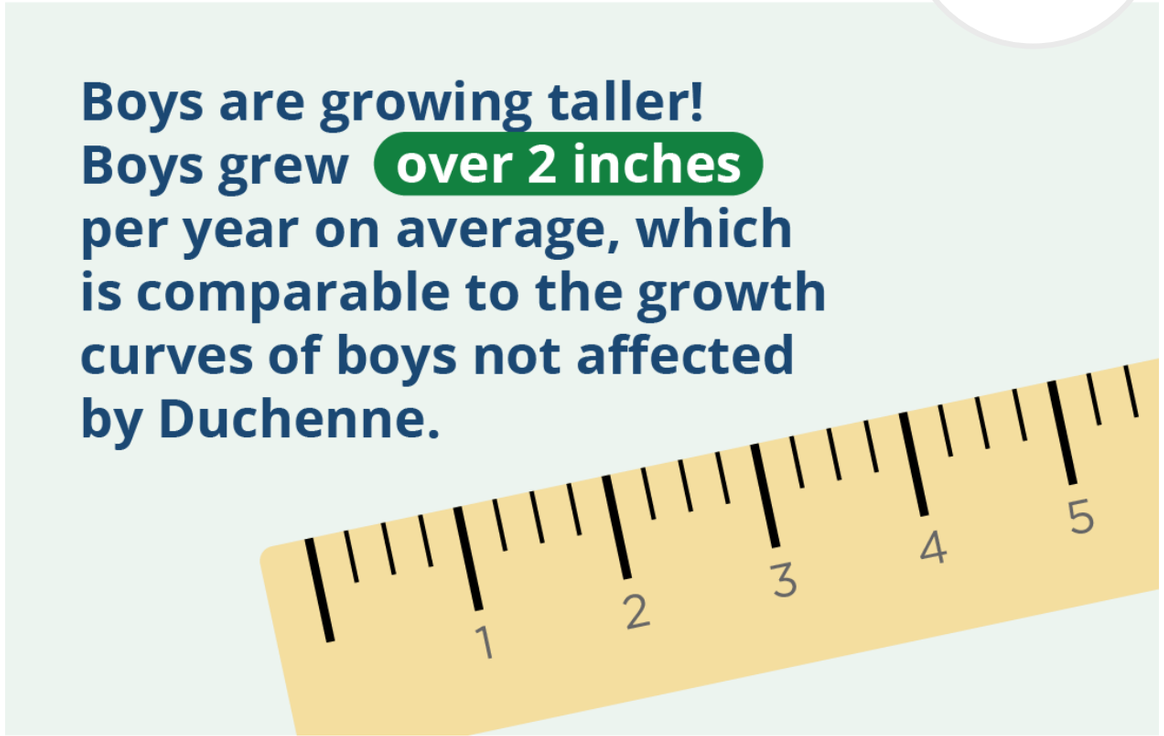

Boys are growing taller!
Boys grew over 2 inches per year on average, which is comparable to the growth curves of boys not affected by Duchenne.

In Phase 2 MoveDMD Trial and Open-Label Extension: Edasalonexent Showed Potential for Cardiac Benefits in DMD

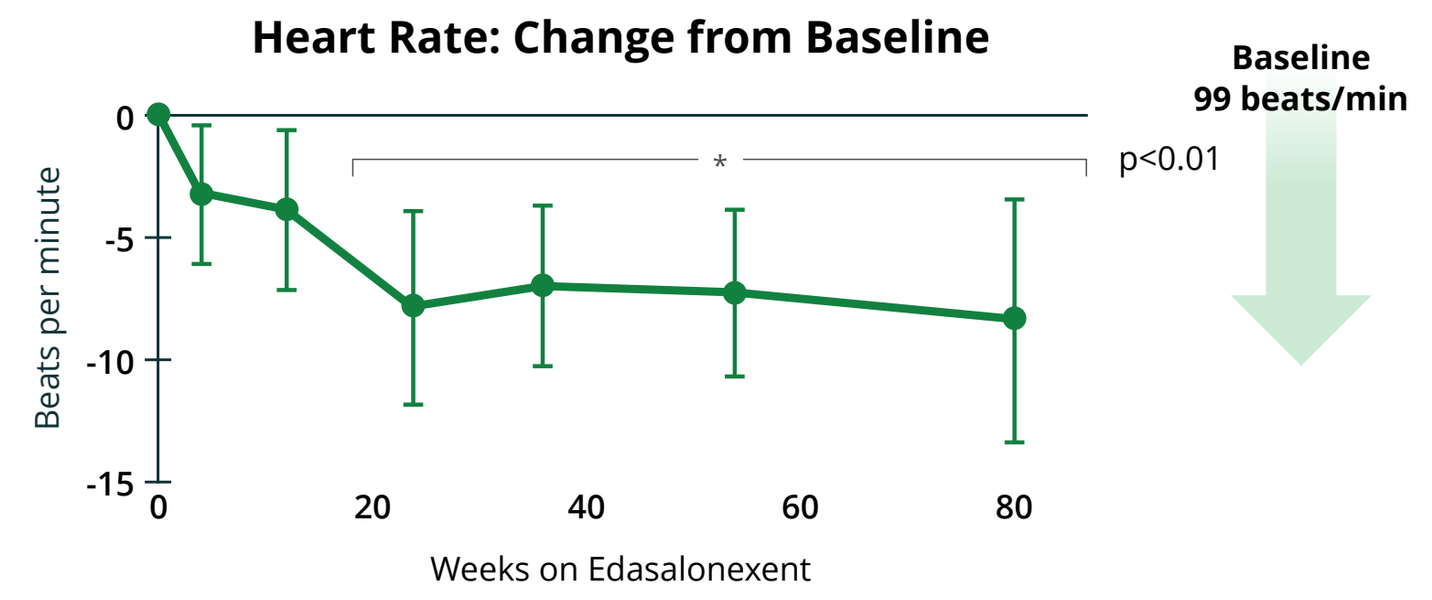

Heart Rate: Change from Baseline
Beats per minute
Baseline 99 beats/min
p<0.01
Weeks on Edasalonexent

- **Elevated resting heart rate is initial manifestation of cardiac disease in DMD**
 - Cardiac failure is a leading cause of mortality in DMD
 - Elevated heart rate triples the risk of cardiomyopathy several years later
- **Edasalonexent analog had positive effects on fibrosis in mdx and GRMD models**
 - Edasalonexent tissue levels in heart > skeletal muscle
- **In MoveDMD trial, mean resting heart rate significantly decreased, approaching age-normative heart rate ~92 beats per minute**

Thomas, et al. *Pediatr Cardiol.* 2012 33(7):1175-9.
Hammers, et al. *JCI Insight* 2016 1(21): e90341
Fleming, et al., *Lancet* 2011 377: 1011-18

Means ± SEM shown;

8

Conclusions & Acknowledgements

Edasalonexent: Potential to Slow Disease Progression for All Those Affected by Duchenne

- Being developed as a new oral foundational therapy for all affected by Duchenne, regardless of mutation type, from time of diagnosis throughout their lifetime
- Being developed for treatment alone and potential to be combined with dystrophin-targeted therapies
- In Phase 2 MoveDMD trial and open-label extension, edasalonexent substantially slowed disease progression compared to off-treatment control period
- Edasalonexent is an investigational agent not currently approved in any territory

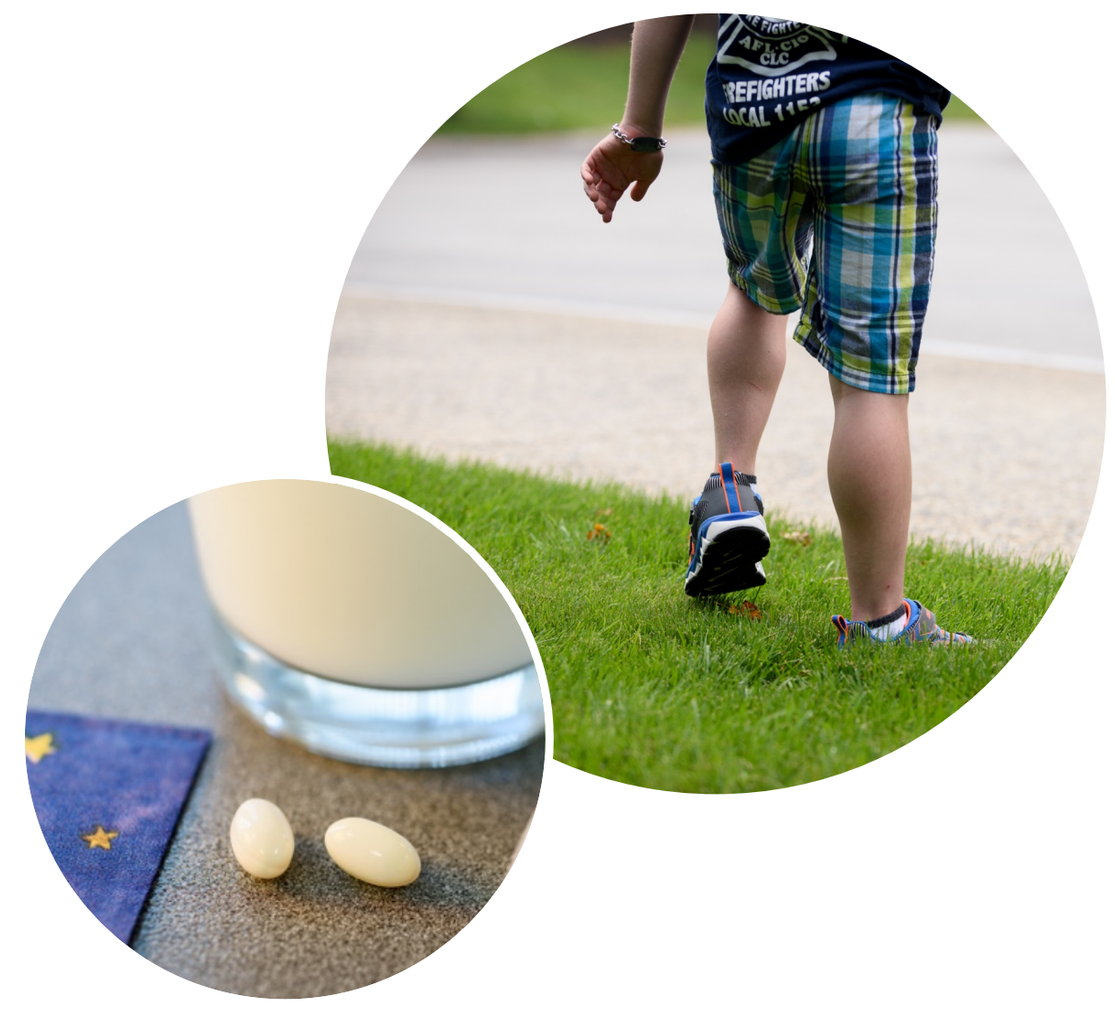

Acknowledgments

We would like to thank patients, families, investigators, site staff, and advocacy organizations.

Thank you to PPMD and the MDA for providing travel support.

EMAIL the team at DMDtrials@catabasis.com

FOLLOW US on social media for frequent updates @CatabasisPharma

SIGN UP to receive our newsletter and information updates on our website

catabasis

Presented at PPMD 25th Annual Conference | June 2019 | Orlando, Florida